Safety and efficacy of sofosbuvir containing regimens for hepatitis C: Community treatment of a real world population with advanced liver disease

EASL The Home of Hepatology
50th THE INTERNATIONAL LIVER CONGRESS 2015
APRIL 22-26, VIENNA, AUSTRIA

D. Ouzan (1), T. Fontanges (2), G. Penaranda (3), P. Delasalle (4), M. Antoni (5), S. Baesjou (6), S. Bresson Hadni (7), P. Cerdan (8), N. Boyer (9), C. Castelnau (10), S. Beorchia (11), J-P. Jacques (12), H. Joly (1), C. Bonny (13), P. Toulemonde (10), J. Liautard (14), B. Hanslik (15)
(1) Saint Laurent du Var; (2) Bourgoin-Jallieu; (3) Marseille; (4) Grasse; (5) Orange; (6) La Rochelle; (7) Besançon; (8) Nîmes; (9) Nanterre; (10) Paris; (11) Lyon; (12) Toulouse; (13) Beaumont; (14) Saint Jean de Vedas; (15) Montpellier. Et la Commission Hépatologie du CREGG

BACKGROUND

The use of direct-acting antivirals such as Sofosbuvir (SOF), Simeprevir (SIME) and Daclatasvir (DACLA) created a major paradigm shift in the treatment of chronic hepatitis C. The aim of this study was to evaluate the safety and efficacy of DDAs utilized in clinical practice in experienced patients with advanced liver fibrosis

OBJECTIVES

The aim of this study was to evaluate the safety and efficacy of DDAs utilized in clinical practice in experienced patients with advanced liver fibrosis

MATERIALS & METHODS

AVD LIB is a longitudinal observational study of patients treated with DDAs at community medical centers (n=13). Demographic, clinical, adverse events and virological data were collected during treatment and post treatment follow-up. From March to September 2014, 209 patients have started treatment according to ATU French guidelines: (Interferon bases regimen failure and fibrosis F3 or F4) and are included in this study.

RESULTS

Patients characteristics

Mean year : 43.3±13.0 years
Sex: Male (58%) / Female (42%)
HCV Genotype : 1 (70%)
2 (3%)
3 (16%)
4 (11%)
Fibrosis score : F3: 72 (33%)
F4 : 137 (67%) Child A: 120, Child B: 17
Viral load >800 000 UI/ml: (70%)
Naive : 41 (20%)
Treatment failure : 168 (80%)
Interferon intolerant : 73 (35%)
Co infected VIH-VHC: 11 (5%)
Liver Transplant : 7 (3%)
Alcool > 30g/l (12.5%)
Diabetes (11%)
HTA (18%)

Treatment

- Sofosbuvir plus ribavirin alone (n=20) 12 (n=3) or 24 weeks (n=17)
- Sofosbuvir plus pegylated interferon/ribavirin 12 weeks (n=44)
- Sofosbuvir plus simeprevir, with (5) or without ribavirin (49) 12 weeks (n=54)
- Sofosbuvir plus daclatasvir, with (15) or without ribavirin (76) 12 (n=15) or 24 weeks (n=76)

Patients (n=209)

Lost of follow up (n=2)
Treatment Discontinued (n=0)

SVR 12 not available (n=76)

SVR 12 Available (n=131)

SVR 12 Achieved (n=114)

SVR 12 not Achieved (n=17)

Treatment distribution by HCV genotype

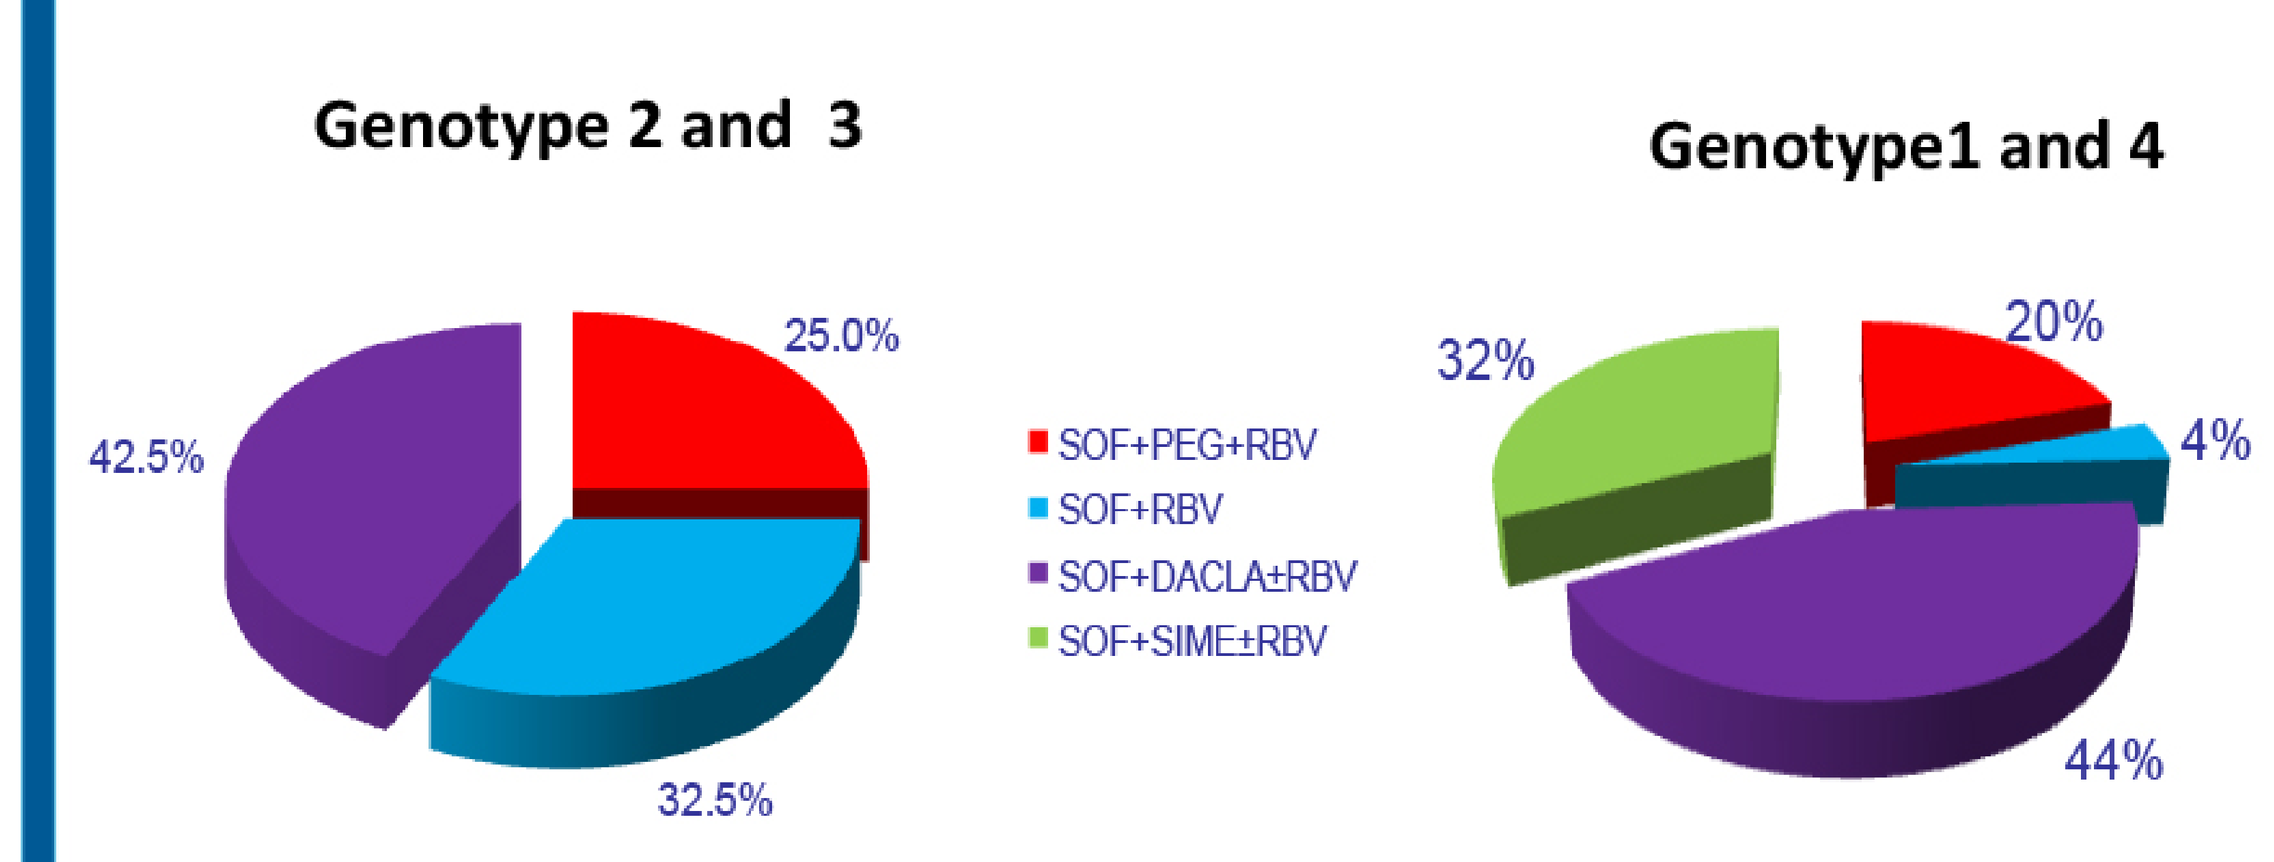
Genotype 2 and 3
25.0%
42.5%
32.5%
SOF+PEG+RBV
SOF+RBV
SOF+DACLA±RBV
SOF+SIME±RBV
Genotype1 and 4
32%
20%
4%
44%

SVR12 score in 131 patients according to genotype

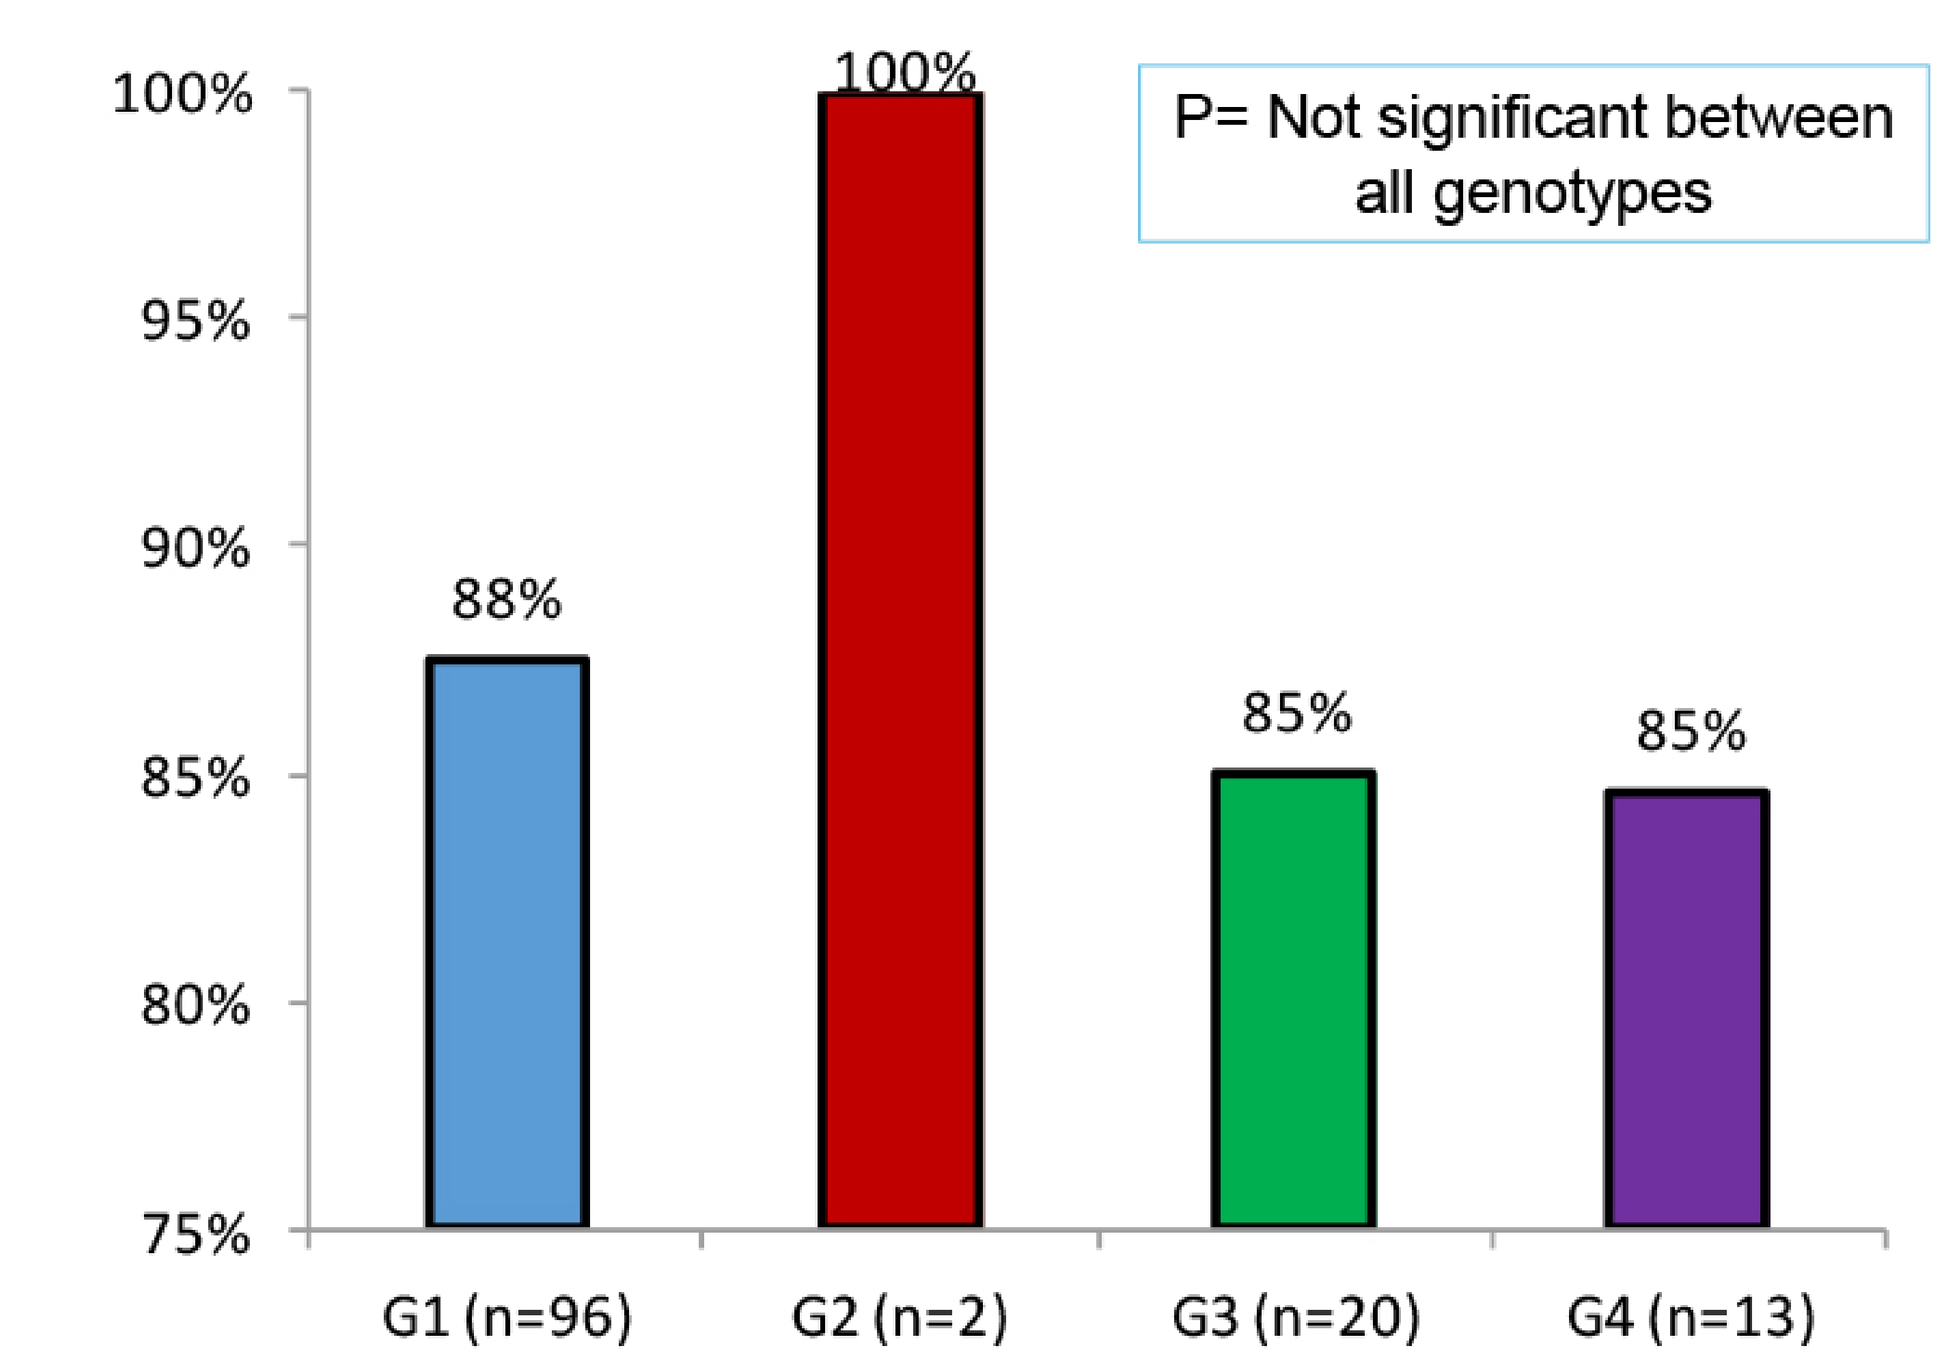
P= Not significant between all genotypes
100%
95%
90%
85%
80%
75%
88%
100%
85%
85%
G1 (n=96)
G2 (n=2)
G3 (n=20)
G4 (n=13)

SVR12 score in 131 patients according to treatment

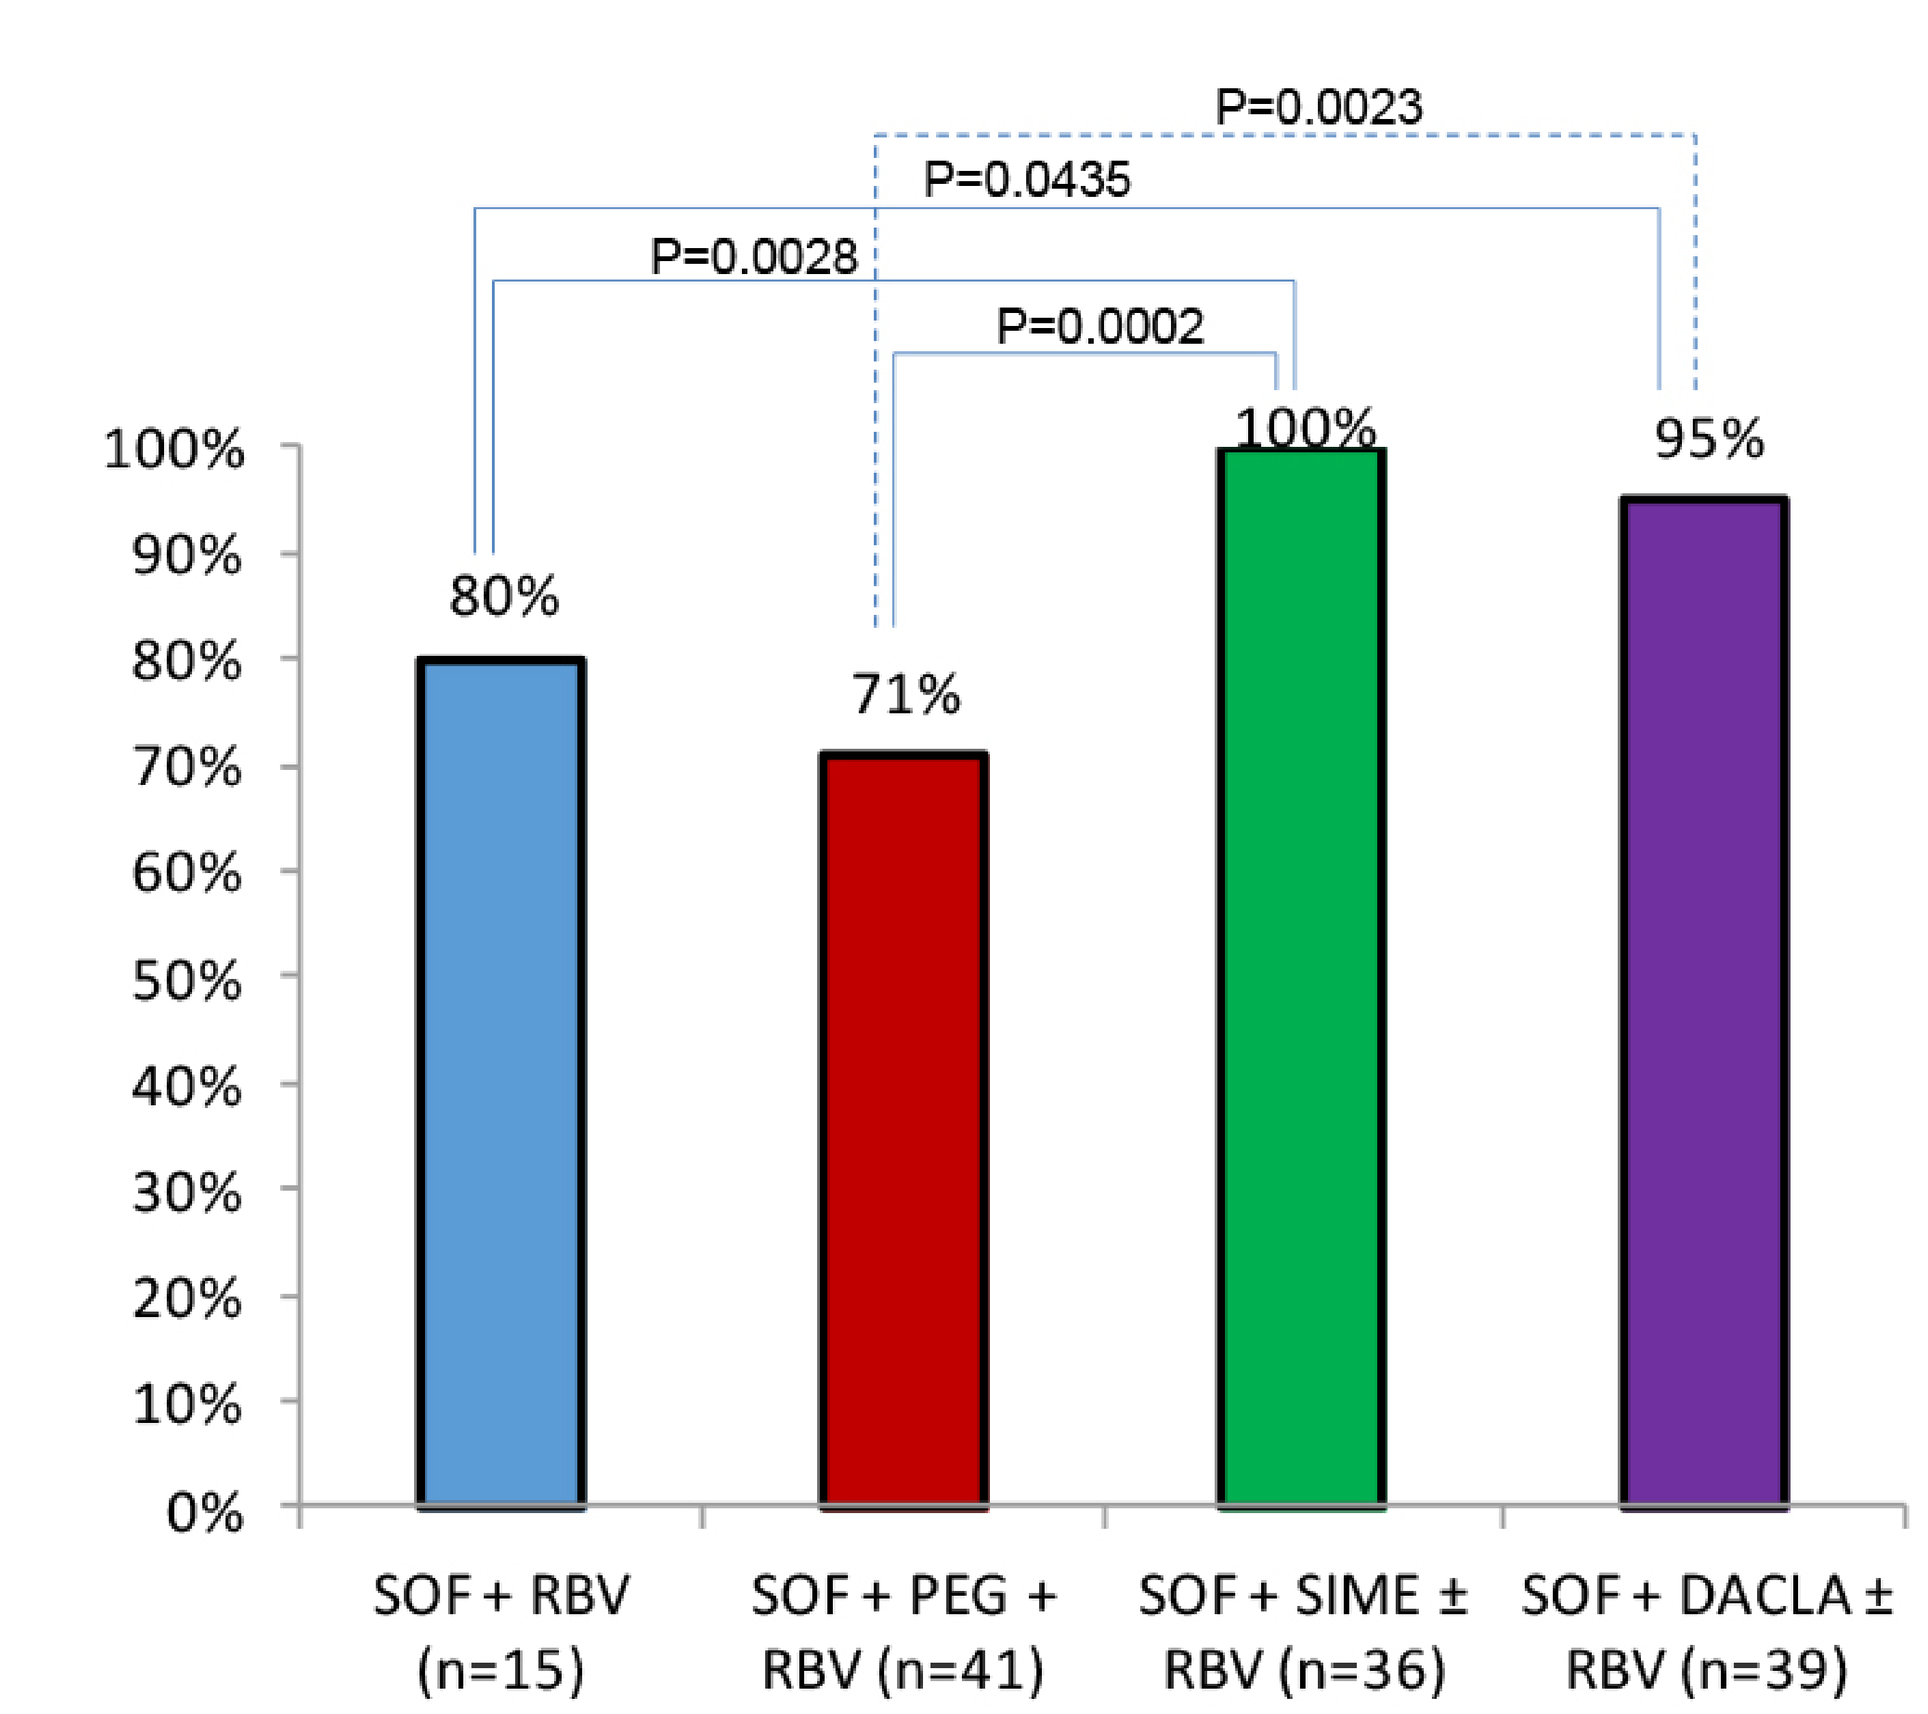
P=0.0023
P=0.0435
P=0.0028
P=0.0002
80%
71%
100%
95%
SOF + RBV (n=15)
SOF + PEG + RBV (n=41)
SOF + SIME ± RBV (n=36)
SOF + DACLA ± RBV (n=39)

SVR12 score in 128 patients according to genotype and treatment

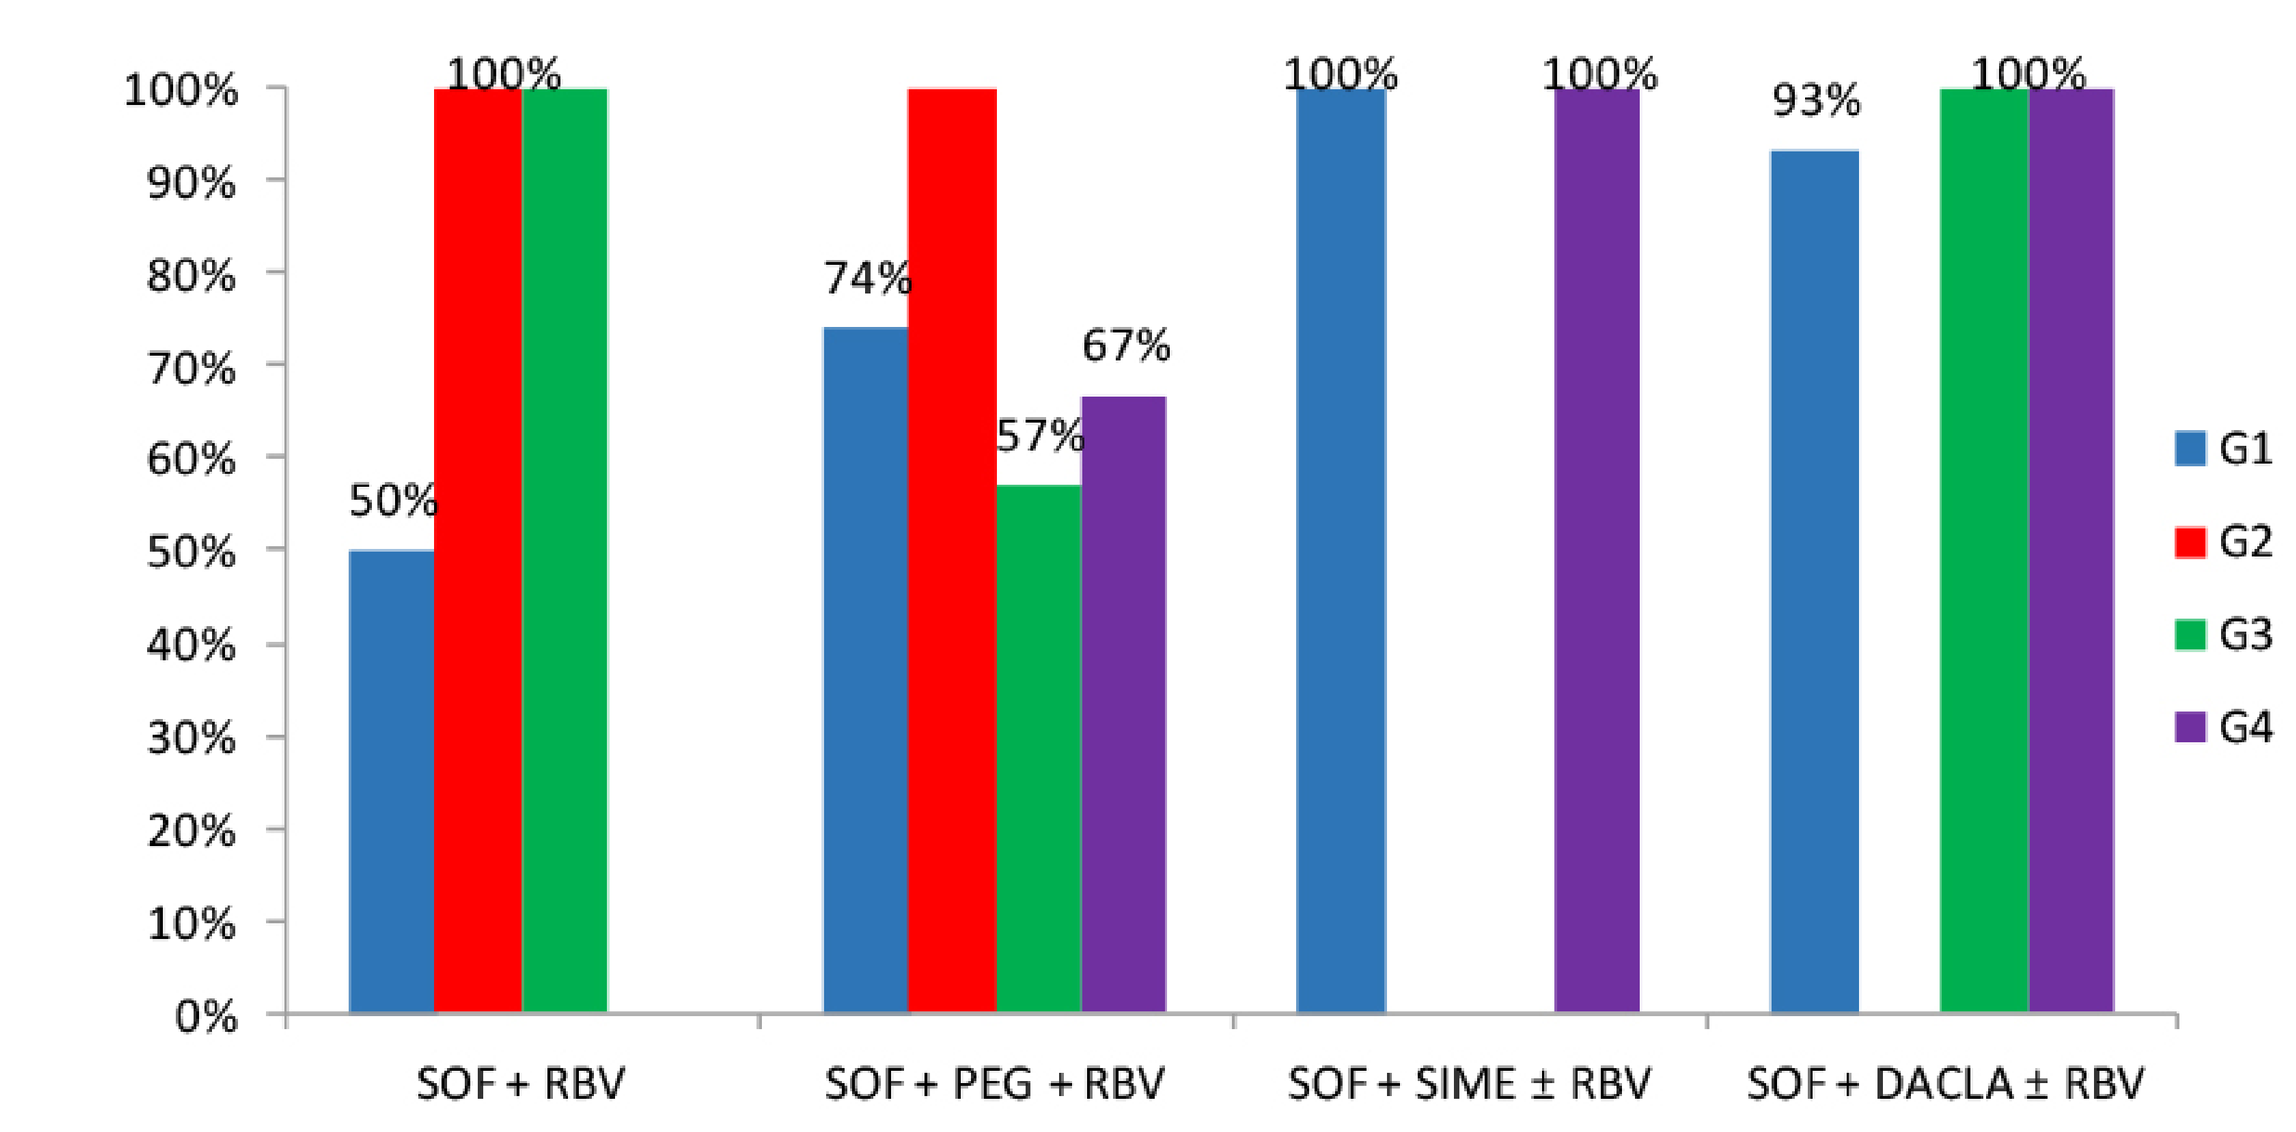
50%
100%
100%
74%
100%
57%
67%
100%
100%
93%
100%
100%
SOF + RBV
SOF + PEG + RBV
SOF + SIME ± RBV
SOF + DACLA ± RBV
G1
G2
G3
G4

SIDE EFFECTS

- Normal life: 105 (50%)
- Fatigue: 65 (31%),
- Headaches: 18 (9%),
- Nausea: 13 (6%),
- Prurit: 6 (3%)

SUMMARY

An examination of real life advanced hepatitis C in community medical practice is underway.
Two interferon-free regimens containing the direct-acting antivirals sofosbuvir plus simeprevir, and sofosbuvir plus daclatasvir led to high sustained virological response rates, and were generally well tolerated. The life of interferon based regimen is coming to the end and replaced by the combination of direct acting antivirals.

Contact Information

Denis OUZAN denis.Ouzan@wanadoo.fr